October 2-6 • Washington, DC • www.idweek.org

IDWeek 2019™ Advancing Science, Improving Care

N° poster

Purified personalized bacteriophages as part of salvage therapy in patients with complex BJI: A 2-year experience in a reference center in France

T. Ferry[1-4] , C. Kolenda[1-3], G. Leboucher[1-3], C. Batailler[-1-3], C. Fevre[5], S. Lustig[1-3], F. Laurent [1-4], on behalf of the Lyon BJI Study group*

Hospices Civils de Lyon, [2] UCBL1, [3] CRIOAc Lyon, [4]CIRI, [5]Pherecydes Pharma

Aim

To report our experience in treating patients with purified personalized bacteriophages.

Method

Seven purified bacteriophages against S. aureus and/or P. aeruginosa, have been selected and manufactured by a French private company by approaching good manufacturing practice (GMP) standards.

Under the supervision of the French National Agency for Medicines and Health Products Safety (ANSM), we have the ability to treat some patients with complex BJI as compassionate use for salvage therapy with these bacteriophages.

Patients were selected during multidisciplinary meeting. For all patients, S. aureus and/or P. aeruginosa strains responsible for the infections have to be isolated before surgery, and a phagogram (i.e. susceptibility of the strain to each bacteriophage; performed by spot test and killing assay) has to be performed to facilitate the selection of the best bacteriophage mix for the treatment.

The final bacteriophage mix was done just before the surgery, by the hospital pharmacist.

Results

Five patients with complex BJI were treated with purified personalized bacteriophages as part of salvage therapy.
Two patients received anti-*S. aureus*, two patients received anti-*P. aeruginosa*, and one patient received anti-*S. aureus* and anti-*P. aeruginosa* bacteriophages personalized mix, based on the results of the phagogram. One patient had chronic osteomyelitis due to multidrug-resistant *P. aeruginosa*. This patient received four administration before performing a local flap. Locally the outcome was favorable but unfortunately the patient rapidly died due to metastatic lung cancer.

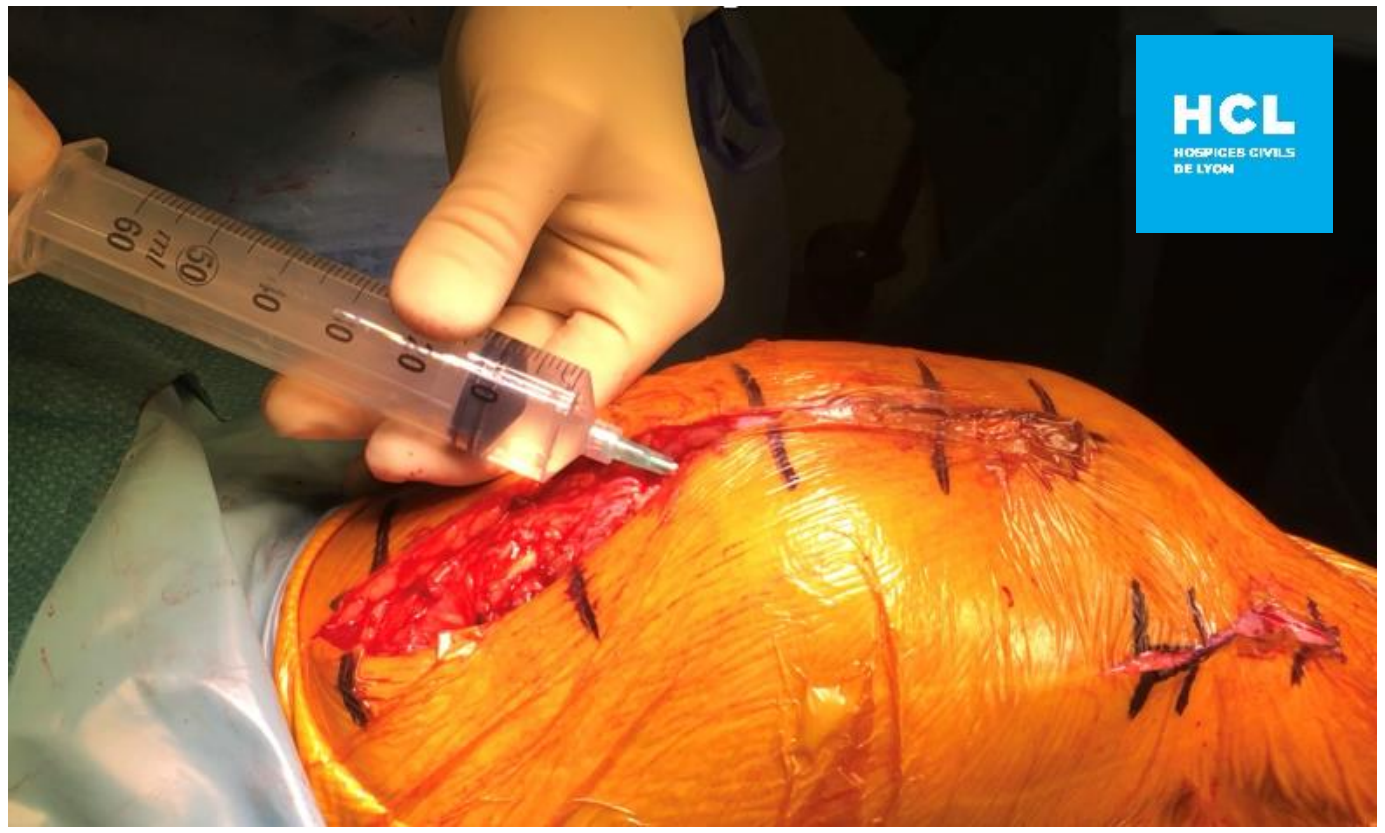
HCL
HOSPICES CIVILS DE LYON

PhagoDAIR concept:

Selected GMP-Bacteriophages

One-shot administration during DAIR in patients with PJI

The four other patients had chronic relapsing prosthetic joint infection (PJI; 3 knees, 1 hip) treated by debridement and implant retention (DAIR), local administration of the selected mix of bacteriophages, and antibiotics (PhagoDAIR concept).
For one of them, the mix of bacteriophages was prepared in a specific hydrogel, as the patient had a large-resection infected knee prosthesis requiring a large free flap performed in the same surgery. Unfortunately, the patient developed hematoma, with superinfection due to Enterobacteriaceae and fistula. The patient with hip PJI was treated 6 months, and experienced hematogenous Enterobacteriaceae 2 years later, requiring a new DAIR. The last two patients had relapsing knee PJI, both was treated with DAIR, including DAIR under arthroscopy, with a favorable outcome under oral suppressive antibiotherapy (6 and 17 months of follow-up).

Conclusions

Purified personalized bacteriophages as part of salvage therapy is promising in patients with complex BJI. For patient with relapsing PJI, combination of DAIR (including under arthroscopy), intraarticular administration of bacteriophage mix and suppressive antibiotherapy will be evaluable in a clinical trial.

* Lyon BJI study group

Lyon Bone and Joint Infection Study Group:

Coordinator: *Tristan Ferry*; **Infectious Diseases Specialists** – *Tristan Ferry, Florent Valour, Thomas Perpoint, Patrick Miailhes, Florence Ader, Sandrine Roux, Agathe Becker, Claire Triffault-Fillit, Anne Conrad, Cécile Pouderoux, Marie-Elodie Langlois, Marielle Perry, Fatiha Daoud, Johanna Lippman, Evelyne Braun, Christian Chidiac*; **Surgeons** – *Sébastien Lustig, Elvire Servien, Cécile Batailler, Romain Gaillard, Stanislas Gunst, Julien Roger, Charles Fiquet, Michel Henri Fessy, Anthony Viste, Philippe Chaudier, Jean Luc Besse, Lucie Louboutin, Gaël Gaudin, Tanguy Ledru, Adrien Van Haecke, Quentin Ode, Marcelle Mercier, Florie Alech-Tournier, Sébastien Martres, Franck Trouillet, Cédric Barrey, Emmanuel Jouanneau, Timothée Jacquesson, Ali Mojallal, Fabien Boucher, Hristo Shipkov, Joseph Chateau, Philippe Céruse, Carine Fuchsmann, Arnaud Gleizal*; **Anesthesiologists** – *Frédéric Aubrun, Mikhail Dziadzko, Caroline Macabéo*; **Microbiologists** – *Frederic Laurent, Laetitia Beraut, Céline Dupieux, Camille Kolenda, Jérôme Josse, Claude-Alexandre Gustave*; **Imaging** – *Fabien Craighero, Loic Boussel, Jean-Baptiste Pialat*; **Nuclear Medicine** – *Isabelle Morelec, Marc Janier, Francesco Giammarile*; **PK/PD specialists** – *Michel Tod, Marie-Claude Gagnieu, Sylvain Goutelle*; **Clinical research assistant and database manager**– *Eugénie Mabrut*

HCL HOSPICES CIVILS DE LYON

Lyon 1

ciri Centre International de Recherche en Infectiologie

@FerryLyon
@CrioacLyon

http://www.crioac-lyon.fr

CRIOAc LYON