P102

Salvage 'DAIR' (debridement, antibiotics and implant retention) with local injection of a selected mix of bacteriophages in an elderly patient with relapsing *S. aureus* prosthetic-joint infection

Tristan Ferry,[1-4] * Gilles Leboucher,[1] Cindy Fevre,[5] Yannick Herry,[1,3] Anne Conrad,[1-4] Jérôme Josse, [1-3], Cécile Batailler,[1,3] Christian Chidiac,[1-4] Mathieu Medina,[5] S. Lustig,[1-3] Frédéric Laurent,[1-4] on behalf of the Lyon BJI Study group

[1] Hospices Civils de Lyon, [2] UCBL1, 3 CRIOAc Lyon, [4] CIRI, [5] Pherecydes Pharma

Aim

Bacteriophages have been firstly described in 1917 and remained a popular treatment in Eastern Europe for patients with osteomyelitis. As their production in such countries currently does not follow the European good manufacturing practices (GMP), bacteriophages are never used in patients with PJI, especially due to the risk of pyrogenicity.

Method

A 80-year-old obese woman with a 5 years history of PJI of the right hip without loosening (figure A) requiring suppressive antimicrobial therapy relapsed. Facing suspicion of relapse, the antibiotic was stopped. A purulent discharge appeared with a painful hip (figure B). *S. aureus* grew in culture from the swab of the pus. Pherecydes Pharma (Romainville, France) selected and manufactured (by approaching GMP standards) three bacteriophages against *S. aureus* from its library according to their broad and complementary spectrum, under the supervision of the French National Agency for Medicines and Health Products Safety (ANSM). The DAIR procedure (figure C) revealed pus in contact with the prosthesis. Just before joint closure, the bacteriophage mix was injected into the joint (figure D). Peroperative samples confirmed MSSA in culture but *E. faecalis* (susceptible to amoxicillin) and *S. lugdunensis* (susceptible to all antibiotics, including penicillin) were also detected. The “phagogram” i.e. a measure of the activity of the selected bacteriophages on the *S. aureus* strain that grew preoperatively was done retrospectively. Efficiency of each bacteriophage was tested using efficiency of plating and killing assays.

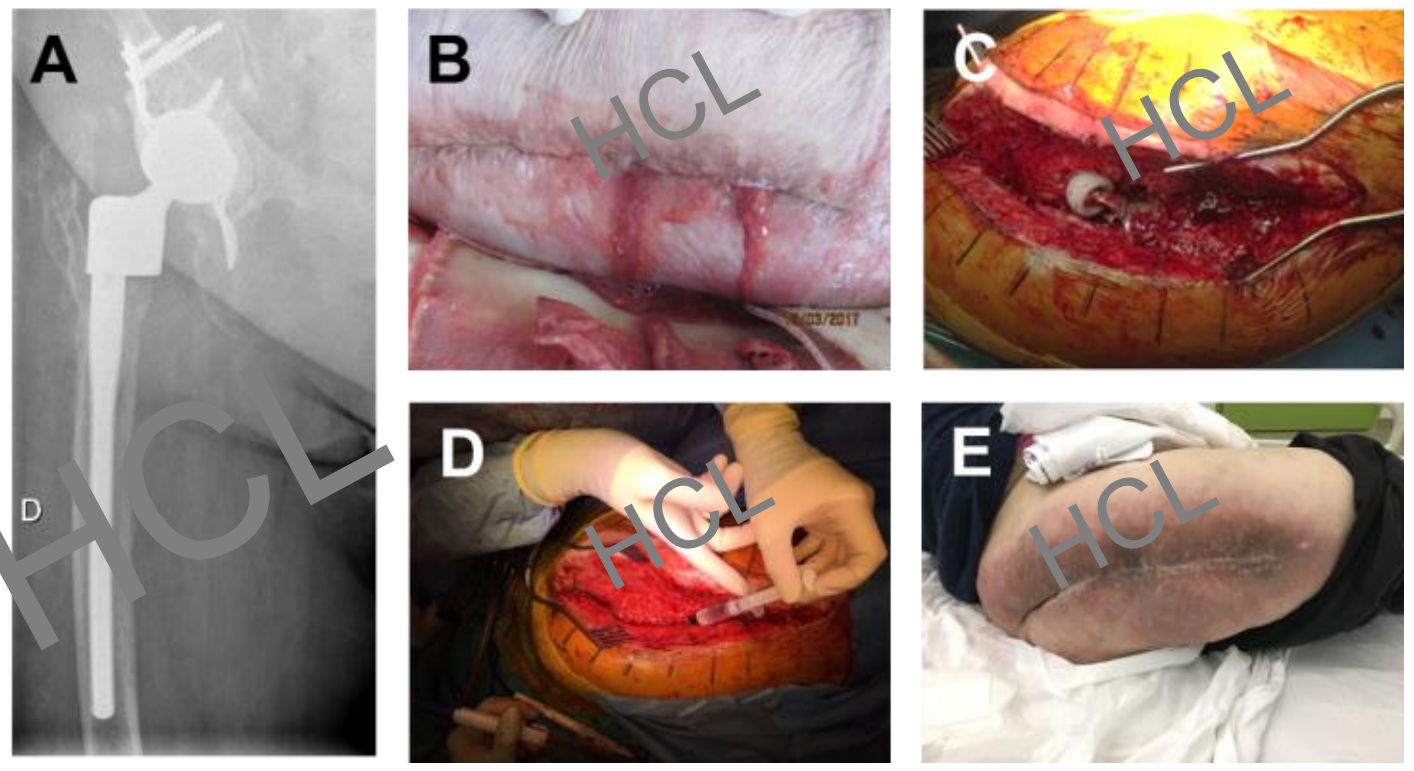

A
B
C
D
E
HCL

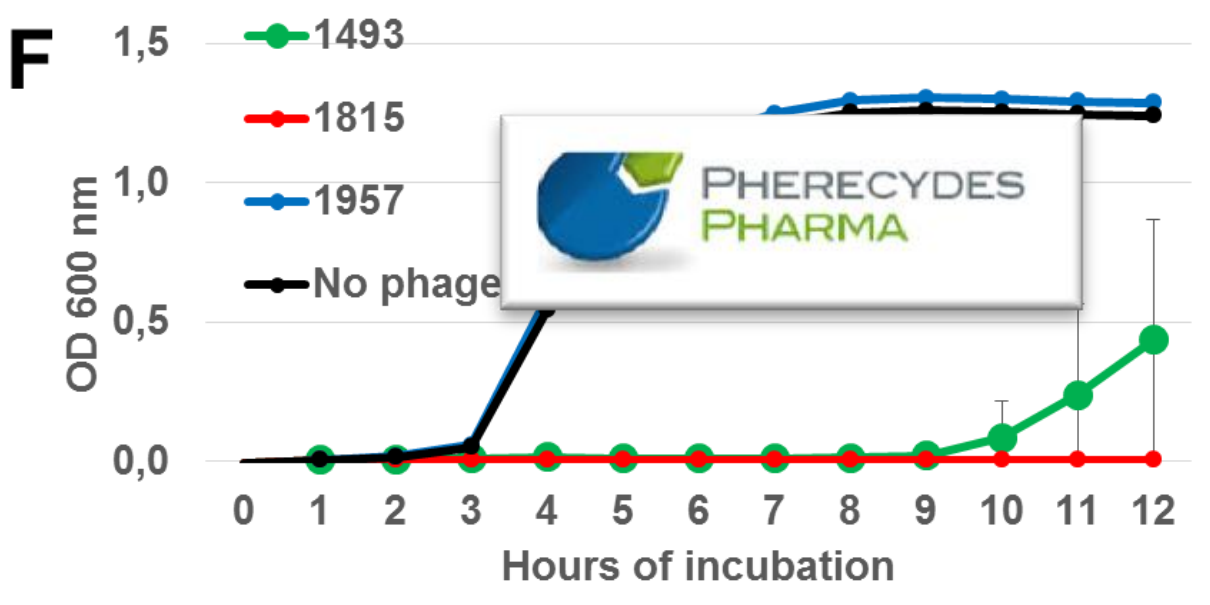

F
1493
1815
1957
No phage
PHERECYDES PHARMA
OD 600 nm
1,5
1,0
0,5
0,0
0 1 2 3 4 5 6 7 8 9 10 11 12
Hours of incubation

Results

The patient received daptomycin after the surgery, then oral treatment (amoxicillin and clindamycin) and finally only amoxicillin, to target *E. faecalis* and *S. lugdunensis* as suppressive antimicrobial therapy. During the follow-up, a new DAIR procedure was performed for a hematogenous *C. koseri* acute hip infection (*S. aureus* did not grow from peroperative cultures) treated with ciprofloxacin during 2 months. One year after the bacteriophages injection, still under amoxicillin, the outcome was favorable without any clinical signs of persistent infection (figure E). The phagogram revealed that only 2 of the 3 bacteriophages used were active and effective against this *S. aureus* strain (figure F). In addition, the bacteriophages had no activity against *S. lugdunensis*.

Conclusion

Local injection of a GMP-like bacteriophages mix during 'DAIR' was performed to treat a relapsing *S. aureus* chronic prosthetic-joint infection. This compassionate treatment was safe and associated with a clinical success. Scientific evaluation of the potential clinical benefit of bacteriophages in bone and joint infection is now feasible and required.

Lyon BJI Study group

Coordinator: *Tristan Ferry*; **Infectious Diseases Specialists** – *Tristan Ferry, Florent Valour, Thomas Perpoint, Patrick Miailhes, Florence Ader, Agathe Becker, Sandrine Roux, Claire Triffault-Fillit, Anne Conrad, Alexie Bosch, Marielle Perry, Fatiha Daoud, Johanna Lippman, Evelyne Braun, Christian Chidiac;* **Surgeons** – *Sébastien Lustig, Elvire Servien, Romain Gaillard, Antoine Schneider, Stanislas Gunst, Cécile Batailler, Michel-Henry Fessy, Yannick Herry, Anthony Viste, Philippe Chaudier, Cyril Courtin, Lucie Louboutin, Sébastien Martres, Franck Trouillet, Cédric Barrey, Emmanuel Jouanneau, Timothée Jacquesson, Ali Mojallal, Fabienne Braye, Fabien Boucher, Hristo Shipkov, Joseph Chateau, Philippe Céruse, Carine Fuchsmann, Arnaud Gleizal;* **Anesthesiologists** – *Frédéric Aubrun, Mikhail Dziadzko, Caroline Macabéo*; **Microbiologists** – *Frederic Laurent, Jean-Philippe Rasigade, Laetitia Beraut, Céline Dupieux, Camille Kolenda, Jérôme Josse*; **Imaging** – *Fabien Craighero, Loic Boussel, Jean-Baptiste Pialat*; **Nuclear Medicine** – *Isabelle Morelec, Marc Janier, Francesco Giammarile;* **PK/PD specialists** – *Michel Tod, Marie-Claude Gagnieu, Sylvain Goutelle;* **Clinical research assistant and database manager**– *Eugénie Mabrut*

Visit our website: http://www.crioac-lyon.fr

Lyon 1

Hospices Civils de Lyon

PHERECYDES PHARMA

ciri Centre International de Recherche en Infectiologie

CRIOAc LYON